15th Conference on Retroviruses and Opportunistic Infections
Boston, USA, February 3–6, 2008

Poster 792

Efficacy and Safety of Maraviroc Plus Optimized Background Therapy in Treatment-experienced Patients Infected with CCR5-tropic HIV-1: 48-week Combined Analysis of the MOTIVATE Studies

D Hardy[1], J Reynes[2], I Konourina[3], D Wheeler[4], S Moreno[5], E van der Ryst[3], W Towner[6], A Horban[7], H Mayer[8], and J Goodrich[8]

[1]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [2]University Hospital of Montpellier, Montpellier, France; [3]Pfizer Global Research and Development, Sandwich, UK; [4]Infectious Diseases Physicians, Inc., Annandale, VA, USA; [5]Hospital Ramón y Cajal, Madrid, Spain; [6]Kaiser Permanente, Los Angeles, CA, USA; [7]Szpital Zakazny Centrum Diagnostyki i Terapii AIDS, Warsaw, Poland; [8]Pfizer Global Research and Development, New London, CT, USA

Background

- Maraviroc (Selzentry®/Celsentri®) is an approved CCR5 antagonist with anti-viral activity against CCR5-tropic (R5) HIV-1.[1]
- The efficacy and safety of maraviroc in treatment-experienced patients with R5 virus have been assessed in two randomized, double-blind, placebo-controlled Phase 3 studies; MOTIVATE 1 (USA, Canada) and MOTIVATE 2 (Europe, Australia, USA).[2–5]
- In both studies, maraviroc (QD or BID) + optimized background therapy (OBT) demonstrated significantly greater virologic and immunologic efficacy and a similar safety profile compared to placebo + OBT in the Week 48 primary analysis. These data have been presented independently.[2,3]
- Since the study design, inclusion/exclusion criteria, conduct, monitoring, and statistical analyses of MOTIVATE 1 and MOTIVATE 2 were identical, pooling of data from the two studies was appropriate, resulting in larger sample sizes for subgroup analyses.[6]
- We present the results of a planned analysis of pooled data, at Week 48, from the two MOTIVATE studies.

Methods

- Patients with triple-class drug experience and/or triple-class drug resistance, R5 virus only (Trofile™, Monogram Biosciences), and HIV-1 RNA ≥5,000 copies/mL were randomized in a 1:2:2 ratio to placebo, maraviroc QD, or maraviroc BID, in combination with OBT. [2–5]
- OBT consisted of 3–6 antiretrovirals +/– low-dose ritonavir. Investigational agents (including darunavir, etravirine, and raltegravir) were not used as part of the OBT. Patients receiving a PI (except tipranavir) and/or delavirdine – potent CYP3A4 inhibitors – in their OBT received 150 mg doses of maraviroc; all other patients received 300 mg doses of maraviroc.
- Each study was individually powered to show superiority of either experimental arm compared to the placebo + OBT group.
- The primary endpoint was the change from baseline in log_{10}-transformed plasma HIV-1 RNA levels at 48 weeks. Secondary endpoints included categories of virologic response (percentage of patients with HIV-1 RNA levels <50 and <400 copies/mL), change from baseline in CD4+ cell count, and time to treatment failure.

Results

Study population

- The combined dataset included a total of 1,049 patients who each received at least one dose of maraviroc QD + OBT (n=414), maraviroc BID + OBT (n=426), or placebo + OBT (n=209).
- Demographics and baseline characteristics were similar across the three treatment groups (Table 1).
- The majority of patients had two or fewer potentially active drugs in their OBT regimen according to overall susceptibility score at screening (Table 1).

Table 1: Demographics and baseline characteristics

Total N=1,049	Placebo + OBT N=209	Maraviroc QD + OBT N=414	Maraviroc BID + OBT N=426
Mean age, years (range)	45.7 (29–72)	45.6 (17–75)	46.3 (21–73)
Female, n (%)	24 (11.5)	51 (12.3)	44 (10.3)
White, n (%)	178 (85.2)	336 (81.2)	363 (85.2)
Mean CD4+ count*, cells/mm³	187.2	195.7	189.2
Mean HIV-1 RNA*, log_{10} copies/mL	4.86	4.86	4.85
≤2 active drugs in OBT †, %	66.0	66.0	69.7
Enfuvirtide in OBT †,%	43.5	40.6	42.7
Tipranavir in OBT, %	13.9	15.9	14.8

* Calculated for each patient as the mean of up to three pre-dose assessments (screening, randomization, and baseline) (CD4+ cell count = mean of two pre-dose assessments). † According to overall susceptibility score.
Includes all patients who received at least one dose of study medication; pooled analysis of MOTIVATE 1 and MOTIVATE 2.

Efficacy

- Both maraviroc treatment groups showed greater reductions from baseline in HIV-1 RNA copies/mL at 48 weeks (primary endpoint) compared to placebo + OBT (Table 2).

Table 2: Mean change from baseline HIV-1 RNA at 48 weeks

Total N=1,049	Placebo + OBT N=209	Maraviroc QD + OBT N=414	Maraviroc BID + OBT N=426
Mean change from baseline in HIV-1 RNA copies/ml, log_{10}*	–0.78	–1.68	–1.84
Difference vs placebo group (97.5% confidence interval)		–0.89 (–1.17, –0.62)	–1.05 (–1.33, –0.78)

* HIV-1 RNA value imputed as zero if patient discontinued before 48 weeks.

- For each of the secondary virologic endpoints, including the rates of virologic suppression to <400 and <50 copies/mL at Week 48 (Figure 1), the responses in the maraviroc groups were also significantly better than in the placebo group (*P*<0.0001).

Figure 1: Percentage of patients with HIV-1 RNA suppression (solid lines, HIV-1 RNA <400 copies/mL; dashed lines, HIV-1 RNA <50 copies/mL)

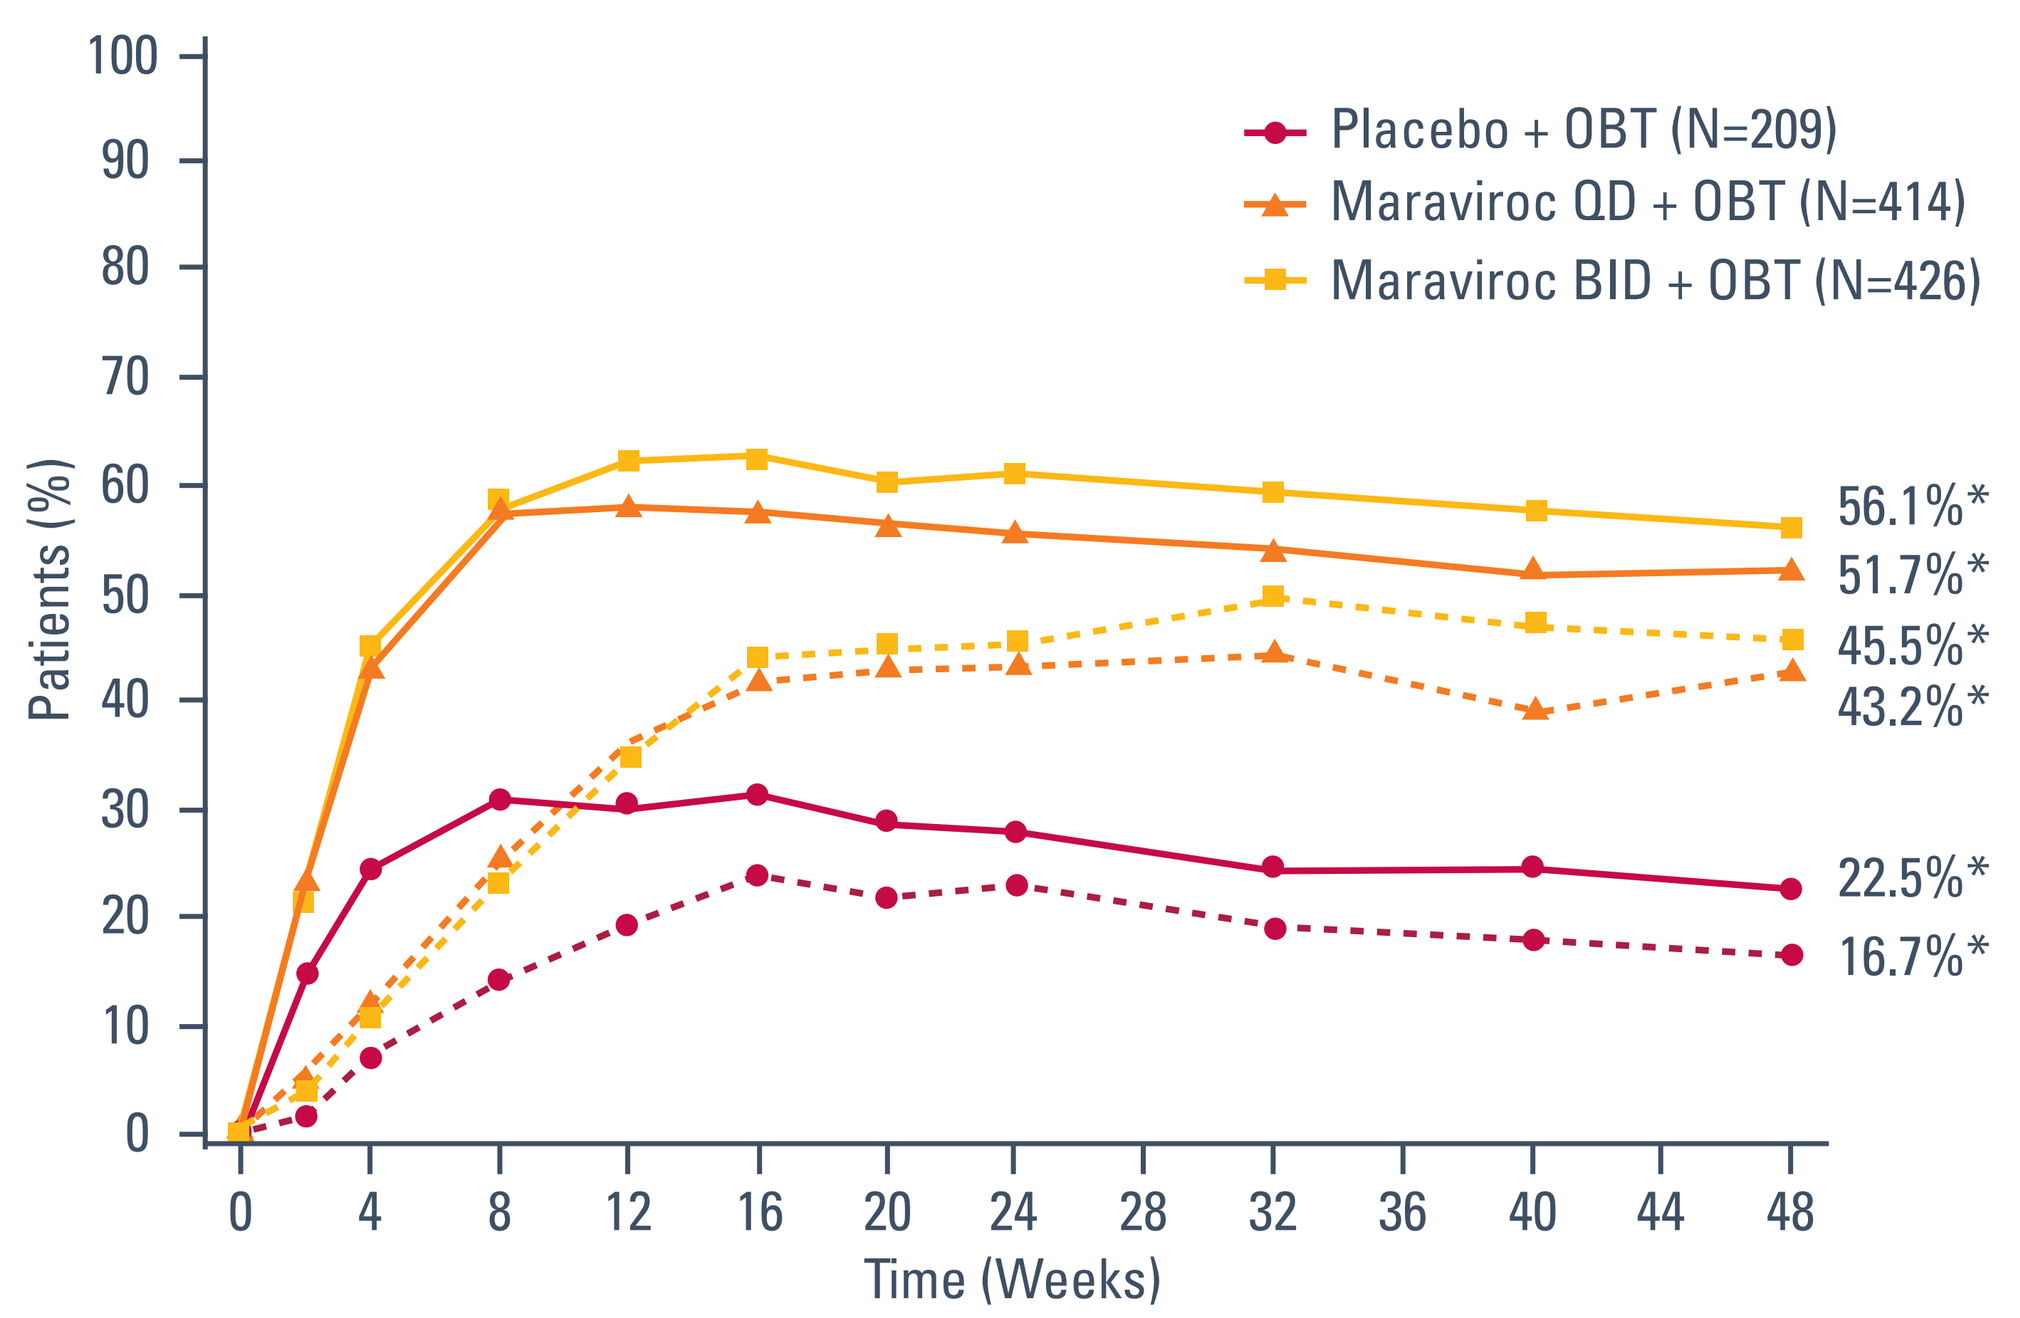
Placebo + OBT (N=209)
Maraviroc QD + OBT (N=414)
Maraviroc BID + OBT (N=426)
Patients (%)
Time (Weeks)
56.1%*
51.7%*
45.5%*
43.2%*
22.5%*
16.7%*

* *P*<0.0001 versus placebo + OBT. HIV-1 RNA value imputed as baseline if missing or if patient discontinued before 48 weeks. Includes all patients who received at least one dose of study medication; pooled analysis of MOTIVATE 1 and MOTIVATE 2.

- CD4+ cell count increases up to 48 weeks were also more favorable in both the maraviroc groups than the placebo group.
 - The mean change from baseline in CD4+ cell count (last-observation-carried-forward approach used to impute missing values) was +61 cells/mm³ in the placebo group, compared to +116 and +124 cells/mm³ in the maraviroc QD and BID groups, respectively.
- Time to treatment failure was similar in the maraviroc QD and BID treatment groups and significantly longer in these groups than in the placebo treatment group (Figure 2).

Figure 2: Kaplan–Meier plot of time to treatment failure

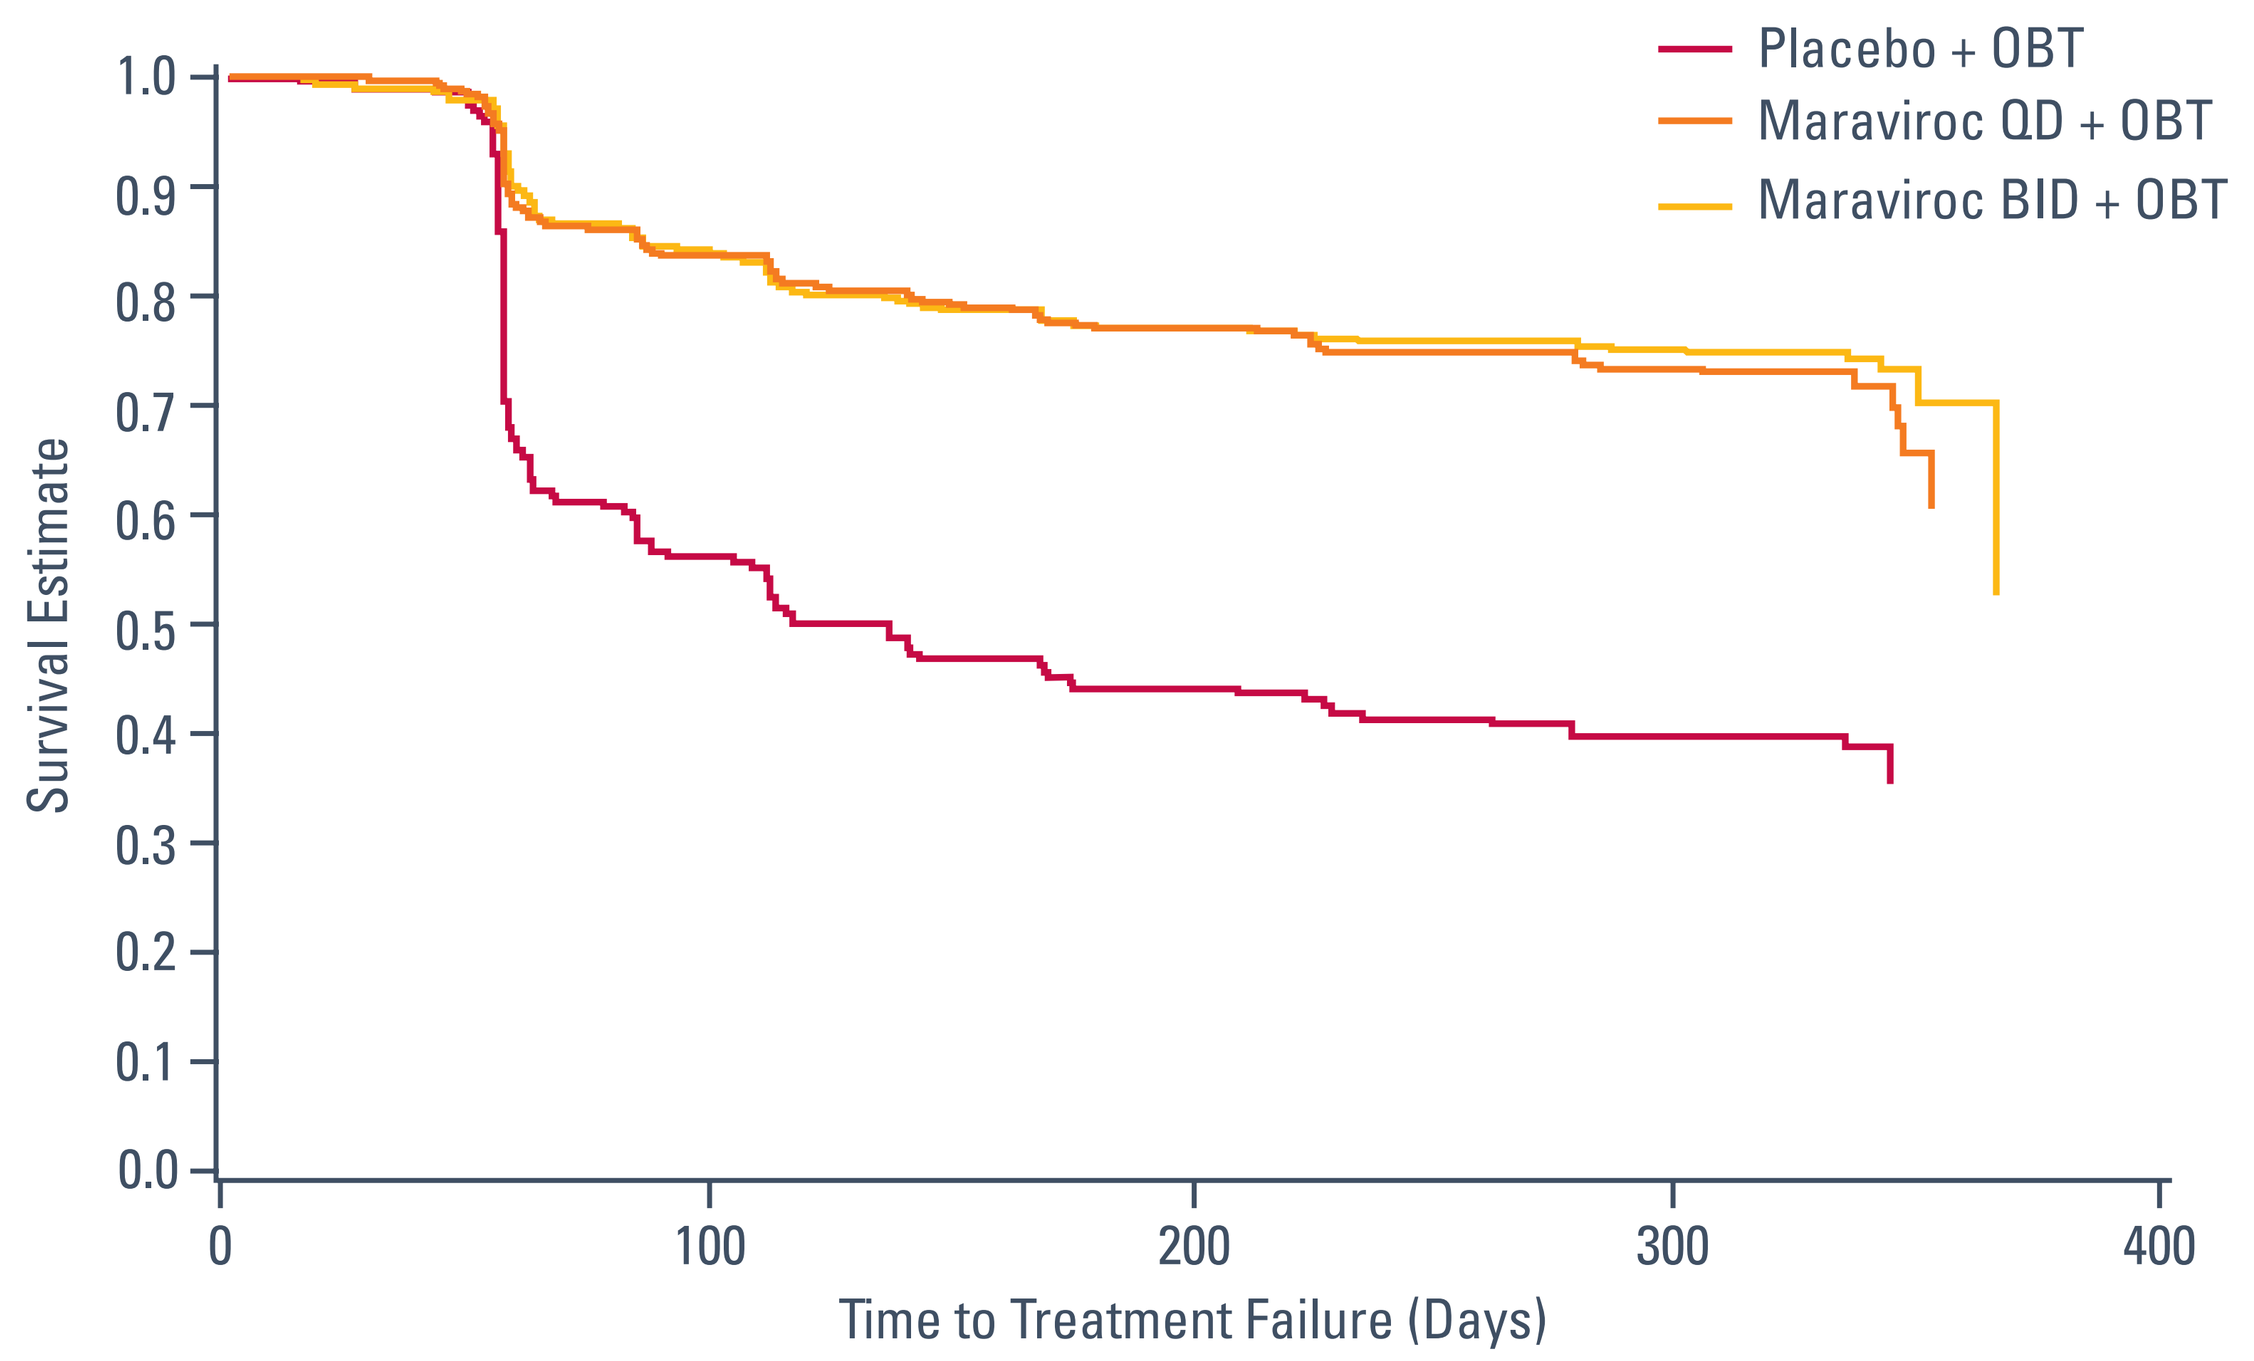
Placebo + OBT
Maraviroc QD + OBT
Maraviroc BID + OBT
Survival Estimate
Time to Treatment Failure (Days)

Includes all patients who received at least one dose of study medication; pooled analysis of MOTIVATE 1 and MOTIVATE 2.

- When treatment response was analyzed by screening viral loads and baseline CD4+ cell counts, the benefit of maraviroc (QD or BID) + OBT over placebo + OBT remained constant across all strata, regardless of whether patients initiated treatment with high viral loads or low CD4+ cell counts (Table 3).

Table 3: Percentage of patients with HIV-1 RNA <50 copies/mL at Week 48 by screening HIV-1 RNA* and by baseline† CD4+ cell count

n/N (%) by subgroup	Placebo + OBT N=209	Maraviroc QD + OBT N=414	Maraviroc BID + OBT N=426
Screening HIV-1 RNA, copies/mL			
<100,000	32/123 (26)	140/238 (59)	142/243 (58)
≥100,000	8/84 (10)	55/170 (32)	61/176 (35)
Baseline CD4+ count, cells/mm³			
<50	1/38 (3)	13/84 (15)	14/85 (16)
50–100	3/25 (12)	19/51 (37)	20/55 (36)
101–200	12/55 (22)	39/95 (41)	59/104 (57)
201–350	13/62 (21)	79/115 (69)	67/116 (58)
>350	10/26 (38)	44/62 (71)	43/59 (73)

* Patients were stratified at time of randomization by screening HIV-1 RNA (< or ≥100,000 copies/mL).
† Baseline CD4+ cell count calculated as the average of all pre-dose measurements.
Includes all treated patients with valid baseline and on-treatment measurements; pooled analysis of MOTIVATE 1 and MOTIVATE 2. Missing values imputed using last observation carried forward.

Safety

- Pooled analysis revealed no new or unique safety findings at Week 48.
- There were no clinically relevant differences in the safety profile between maraviroc (QD and BID) + OBT and placebo + OBT over 48 weeks of treatment (Table 4, Figure 3).
 - This was despite the longer exposure to study treatment in patients in the maraviroc treatment groups (more than 300 patient-years per group) versus the placebo group (111 patient-years) (Table 4), associated with the higher rate of discontinuation in the placebo group.
- Adverse events, serious adverse events, discontinuations due to adverse events, and laboratory abnormalities (including grade 3/4 transaminase elevations) occurred with similar frequency between treatment groups (Table 4).
- The incidence of Category C (AIDS-defining) events was comparable across treatment groups (Table 4).
- The most common adverse events in all treatment groups were diarrhea, nausea, fatigue, and headache (Figure 3), with similar rates for all treatment groups.
- More deaths were recorded in patients receiving maraviroc versus those receiving placebo during the safety analysis reporting period or within 28 days of discontinuing study drug. Deaths were from various causes and none of the deaths were considered to be related to study medication (Table 4).

Figure 3: Percentage of adverse events occurring in ≥5% of patients in any group (unadjusted for study treatment exposure)

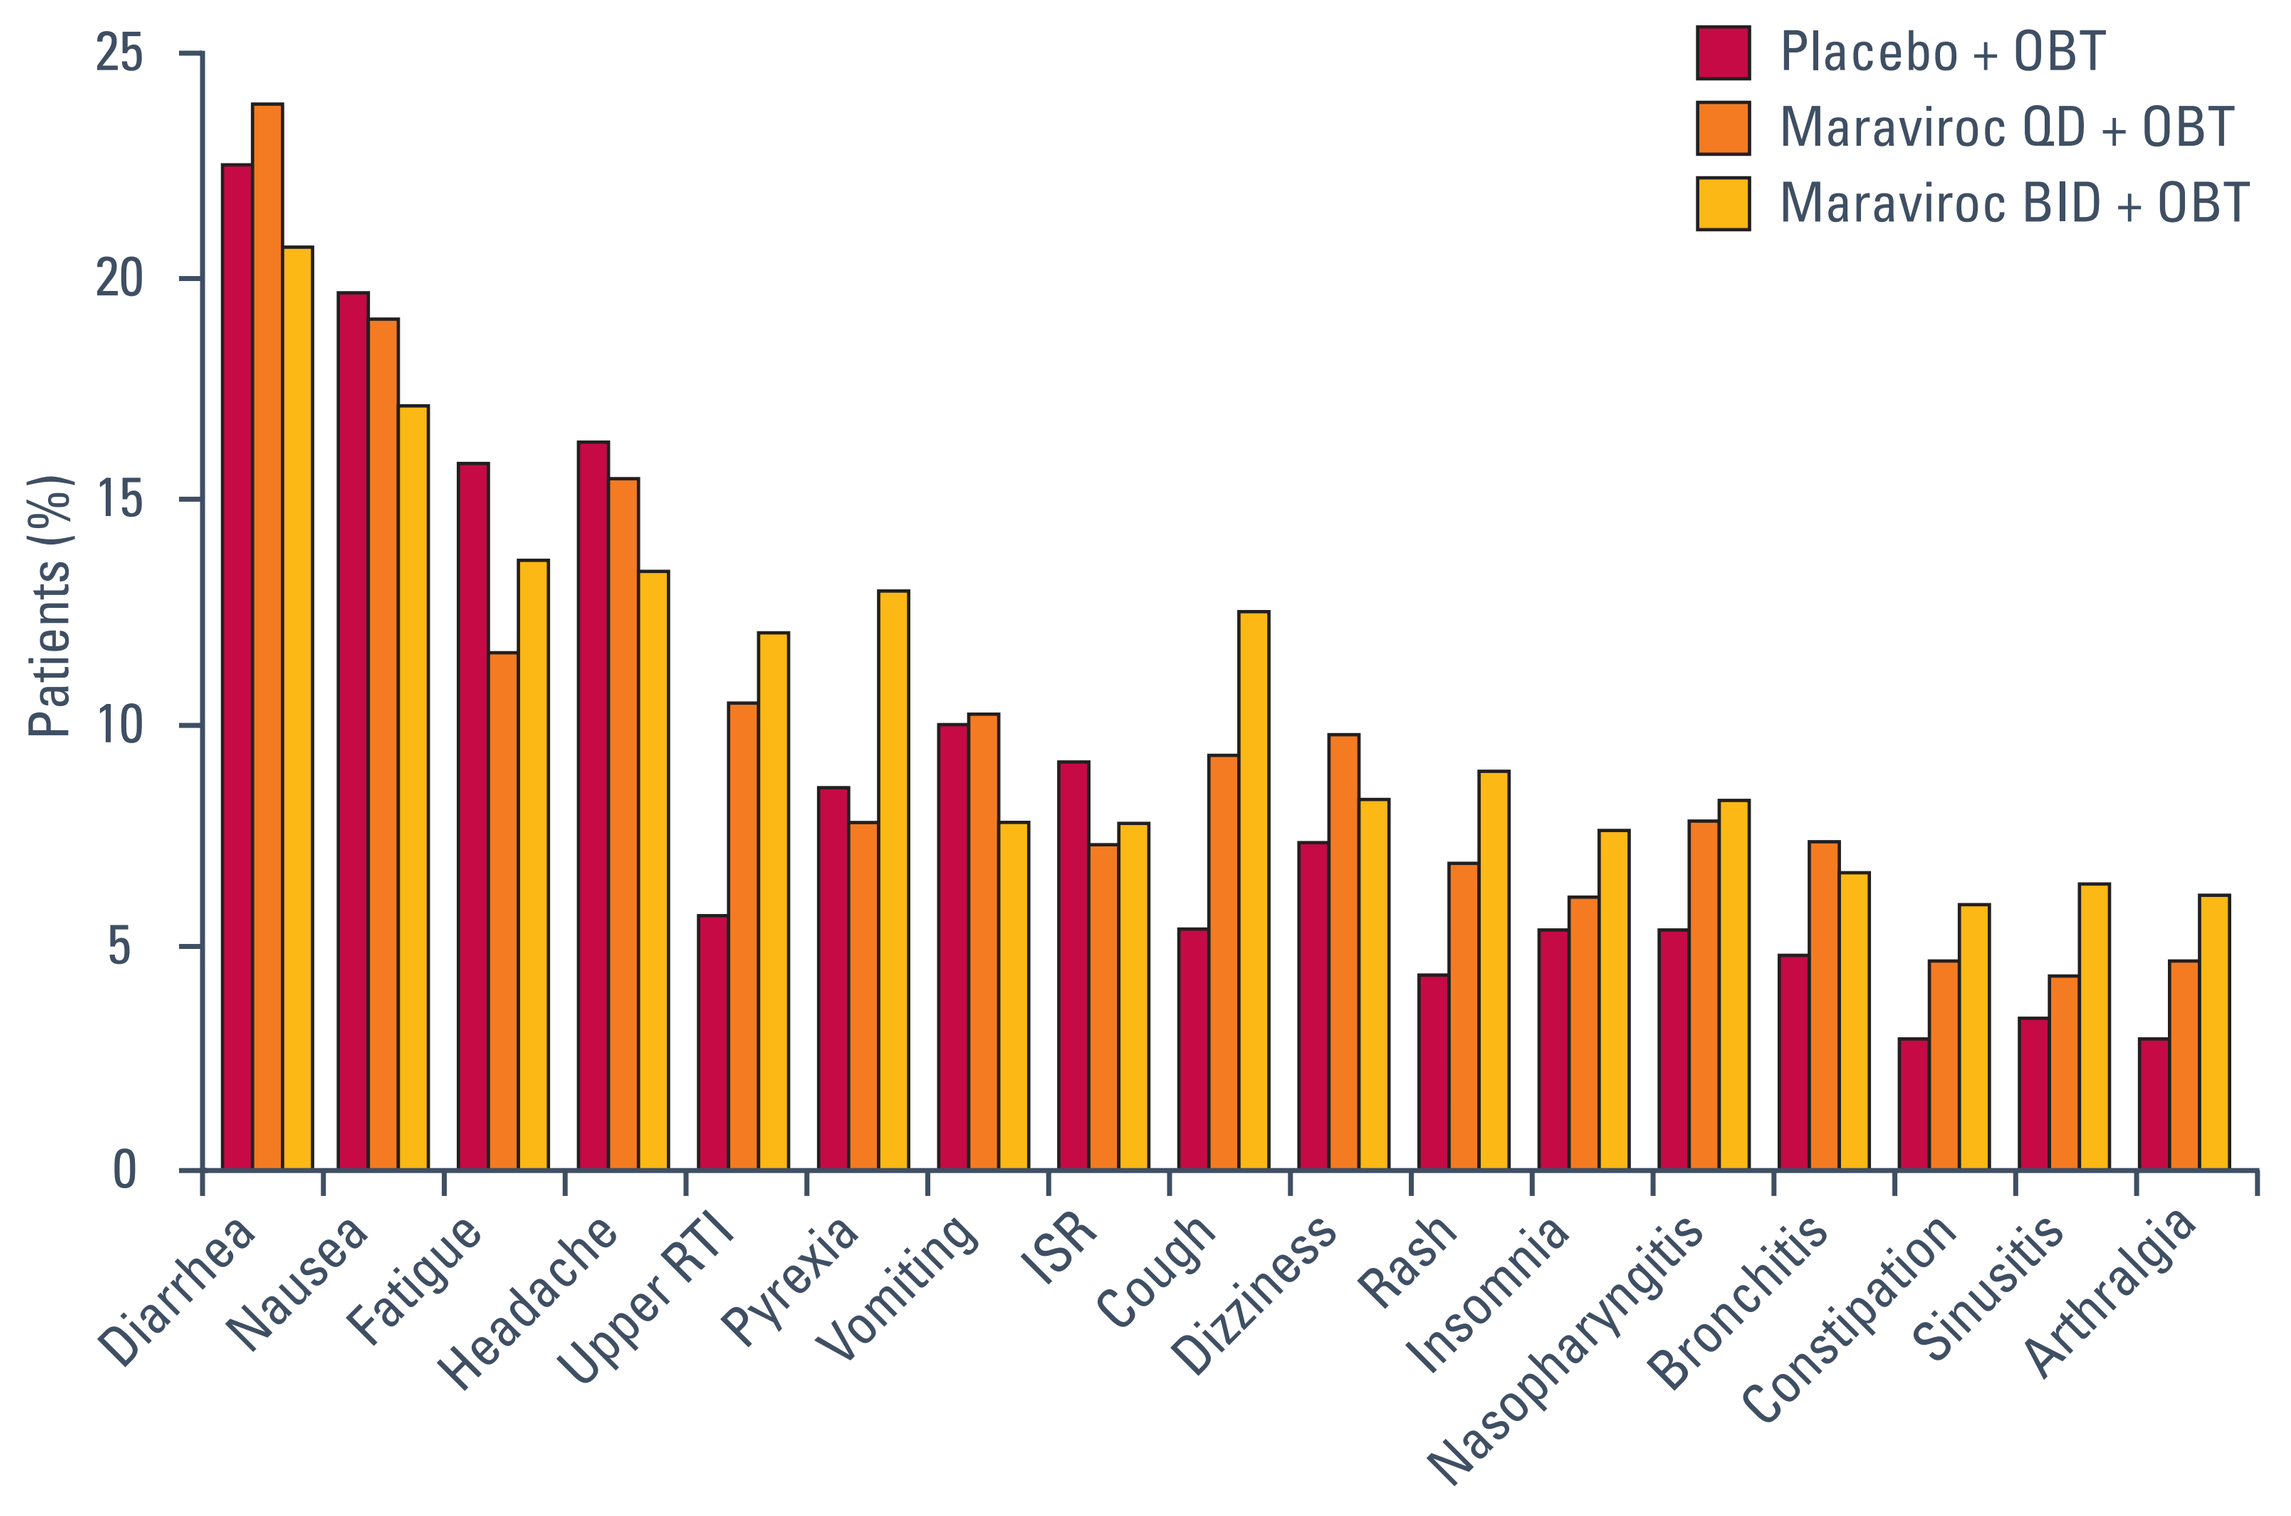
Placebo + OBT
Maraviroc QD + OBT
Maraviroc BID + OBT
Patients (%)
Diarrhea
Nausea
Fatigue
Headache
Upper RTI
Pyrexia
Vomiting
ISR
Cough
Dizziness
Rash
Insomnia
Nasopharyngitis
Bronchitis
Constipation
Sinusitis
Arthralgia

ISR=injection site reaction; RTI=respiratory tract infection. Pooled analysis of MOTIVATE 1 and MOTIVATE 2.

Table 4: Safety analysis (unadjusted for study treatment exposure)

All causalities and severities, n (%)	Placebo + OBT N=209	Maraviroc QD + OBT N=414	Maraviroc BID + OBT N=426
Total exposure to study treatment, patient-years	111	300	309
Treatment-emergent adverse events	177 (84.7)	375 (90.6)	393 (92.3)
Discontinuation due to adverse events	11 (5.3)	24 (5.8)	21 (4.9)
Serious adverse events	35 (16.7)	62 (14.9)	72 (16.9)
Grade 3 adverse events	46 (22.0)	84 (20.3)	104 (24.4)
Grade 4 adverse events	16 (7.7)	37 (8.9)	45 (10.6)
Category C events	16 (7.7)	29 (7.0)	23 (5.4)
Category C malignancies	5 (2.4)	3 (0.7)	4 (0.9)
Non-HIV malignancies	11 (5.3)	12 (2.9)	19 (4.5)
Deaths*	2† (1.0)	6 (1.4)	9 (2.1)
Maximum liver function test values without regard to baseline§ (all causalities), n (%)‡			
Aspartate transaminase			
Grade 3: >5–10 x ULN	6 (2.9)	12 (2.9)	14 (3.3)
Grade 4: >10 x ULN	0	4 (1.0)	6 (1.4)
Alanine transaminase			
Grade 3: >5–10 x ULN	6 (2.9)	16 (3.9)	7 (1.7)
Grade 4: >10 x ULN	1 (0.5)	2 (0.5)	4 (1.0)
Total bilirubin			
Grade 3: >2.5–5 x ULN	8 (3.9◊)	28 (6.9)	21 (5.0◊)
Grade 4: >5 x ULN	3 (1.5◊)	5 (1.2)	2 (0.5◊)

* Deaths reported up to 28 days after stopping study drug; no deaths were related to study drug according to investigator.
† One patient received open-label maraviroc prior to death.
‡ N=207, 408, and 421 for placebo, maraviroc QD, and maraviroc BID groups, respectively.
§ Patients with grade 1 and 2 liver function tests could be enrolled in the studies.[7]
◊ Data missing for one patient.
ULN=upper limit of normal. Pooled analysis of MOTIVATE 1 and MOTIVATE 2.

Conclusions

- Maraviroc (QD or BID) + OBT demonstrated significantly greater virologic suppression rates and increases from baseline in CD4+ cell counts at Weeks 24 and 48 compared to placebo + OBT in this combined analysis.
- The benefit of maraviroc + OBT over placebo + OBT was apparent even in subgroups of patients with adverse prognostic factors, such as high screening viral load or low baseline CD4+ cell count.
- Maraviroc + OBT demonstrated a similar safety profile compared to placebo + OBT, despite considerably greater exposure to study treatment in the maraviroc groups.
- There was no increase in AIDS-defining events (infections or malignancies) or non-AIDS-defining malignancies in maraviroc (QD or BID) + OBT versus placebo + OBT.
- No deaths reported during the study or up to 28 days of stopping study drug were considered to be related to study medication.
- In conclusion, these results demonstrate that, in treatment-experienced patients with R5 virus, maraviroc combined with OBT provides sustained antiretroviral efficacy and tolerability through 48 weeks.

References

1. Dorr P, *et al. Antimicrob Agents Chemother* 2005; 49:4721–4732.
2. Lalezari J, *et al. 47th ICAAC* 2007; Presentation H-718a.
3. Fätkenheuer G, *et al. 11th EACS* 2007; Presentation PS3/5.
4. Lalezari J, *et al. 14th CROI* 2007; Presentation 104bLB.
5. Nelson M, *et al. 14th CROI* 2007; Presentation 104aLB.
6. Gulick RM, *et al. 4th IAS* 2007; Presentation WEPEB116LB.
7. Hoepelman IM, *et al. 11th EACS*; Poster LBP7.9/1.

Acknowledgements

Investigators and study site staff
Patients who participated in the studies
Colleagues from Monogram Biosciences: J Whitcomb, E Coakley, C Petropoulos
R Harrigan, BC Centre for Excellence in HIV
Colleagues from Quintiles
Colleagues from Pfizer: S Felstead, A Bullivant, I Oborska, K George, M Westby, K Turner, D Lindell, D Paige, S Nuttall, J Merson, L Kapili, M Dunne, J Sullivan, C Ridgway, N Rajicic, A Parliyan

Pfizer HIV/AIDS